Prevalence and Dynamics of Drug Resistance During Dolutegravir-Containing Treatment in a Pediatric Population Living with HIV-1 (IMPAACT P1093)

Ceejay L. Boyce[1], Marley D. Bishop[1], Sheila M. Styrchak[1], Kevin Knowles[2], Cindy Vavro[3], Rohan Hazra[4], Ellen Townley[5], Robert Nixon[6], Robert A. Smith[6], Geoffrey S. Gottlieb[6,7], Stephen E. Hawes[8], Paul K. Drain[7], Theodore D. Ruel[9], Andrew A. Wiznia[10], Lisa M. Frenkel[1,6,7,11], and the IMPAACT P1093 Team

1 Center for Global Infectious Disease Research, Seattle Children's Research Institute, Seattle, WA, USA; **2** Frontier Science Foundation, Amherst, NY, USA; **3** ViiV Healthcare, Durham, North Carolina, USA; **4** Eunice Kennedy Shriver National Institute of Child Health & Human Development, National Institutes of Health, Bethesda, MD, USA; **5** National Institute of Allergy and Infectious Diseases, National Institutes of Health, Bethesda, MD, USA; **6** Department of Medicine, University of Washington, Seattle, WA, USA; **7** Department of Global Health, University of Washington, Seattle, WA USA; **8** Department of Epidemiology, University of Washington, Seattle, WA USA; **9** University of California San Francisco, San Francisco, California, USA; **10** Jacobi Medical Center, Albert Einstein College of Medicine, Bronx, New York, USA; **11** Departments of Pediatrics and Laboratory Medicine & Pathology, University of Washington, Seattle, WA, USA

Introduction

- IMPAACT P1093 is a phase I/II study of dolutegravir (**DTG**) and optimized background therapy (**OBT**) in children (4 weeks to <18 years) living with HIV-1
- **A secondary objective of the study is to assess changes in HIV-1 genotypes and phenotypes to DTG and OBT in children with and without intermittent viremia or protocol-defined virologic failure**

Methods

- This study included all 181 children enrolled in P1093 from nine countries in Africa, Americas, and Asia
- Protocol-defined virologic failure (**PDVF**) = two consecutive viral loads >400 c/mL after 24 weeks on study
- Intermittent viremia = ≥2 viral loads >400 c/mL after 24 weeks on study
- Plasma HIV RNA genotyping was performed on screen/enrollment specimens for all (N=181) and longitudinal specimens from participants with PDVF/intermittent viremia (N=49) using PacBio sequencing (**Figure 1**)

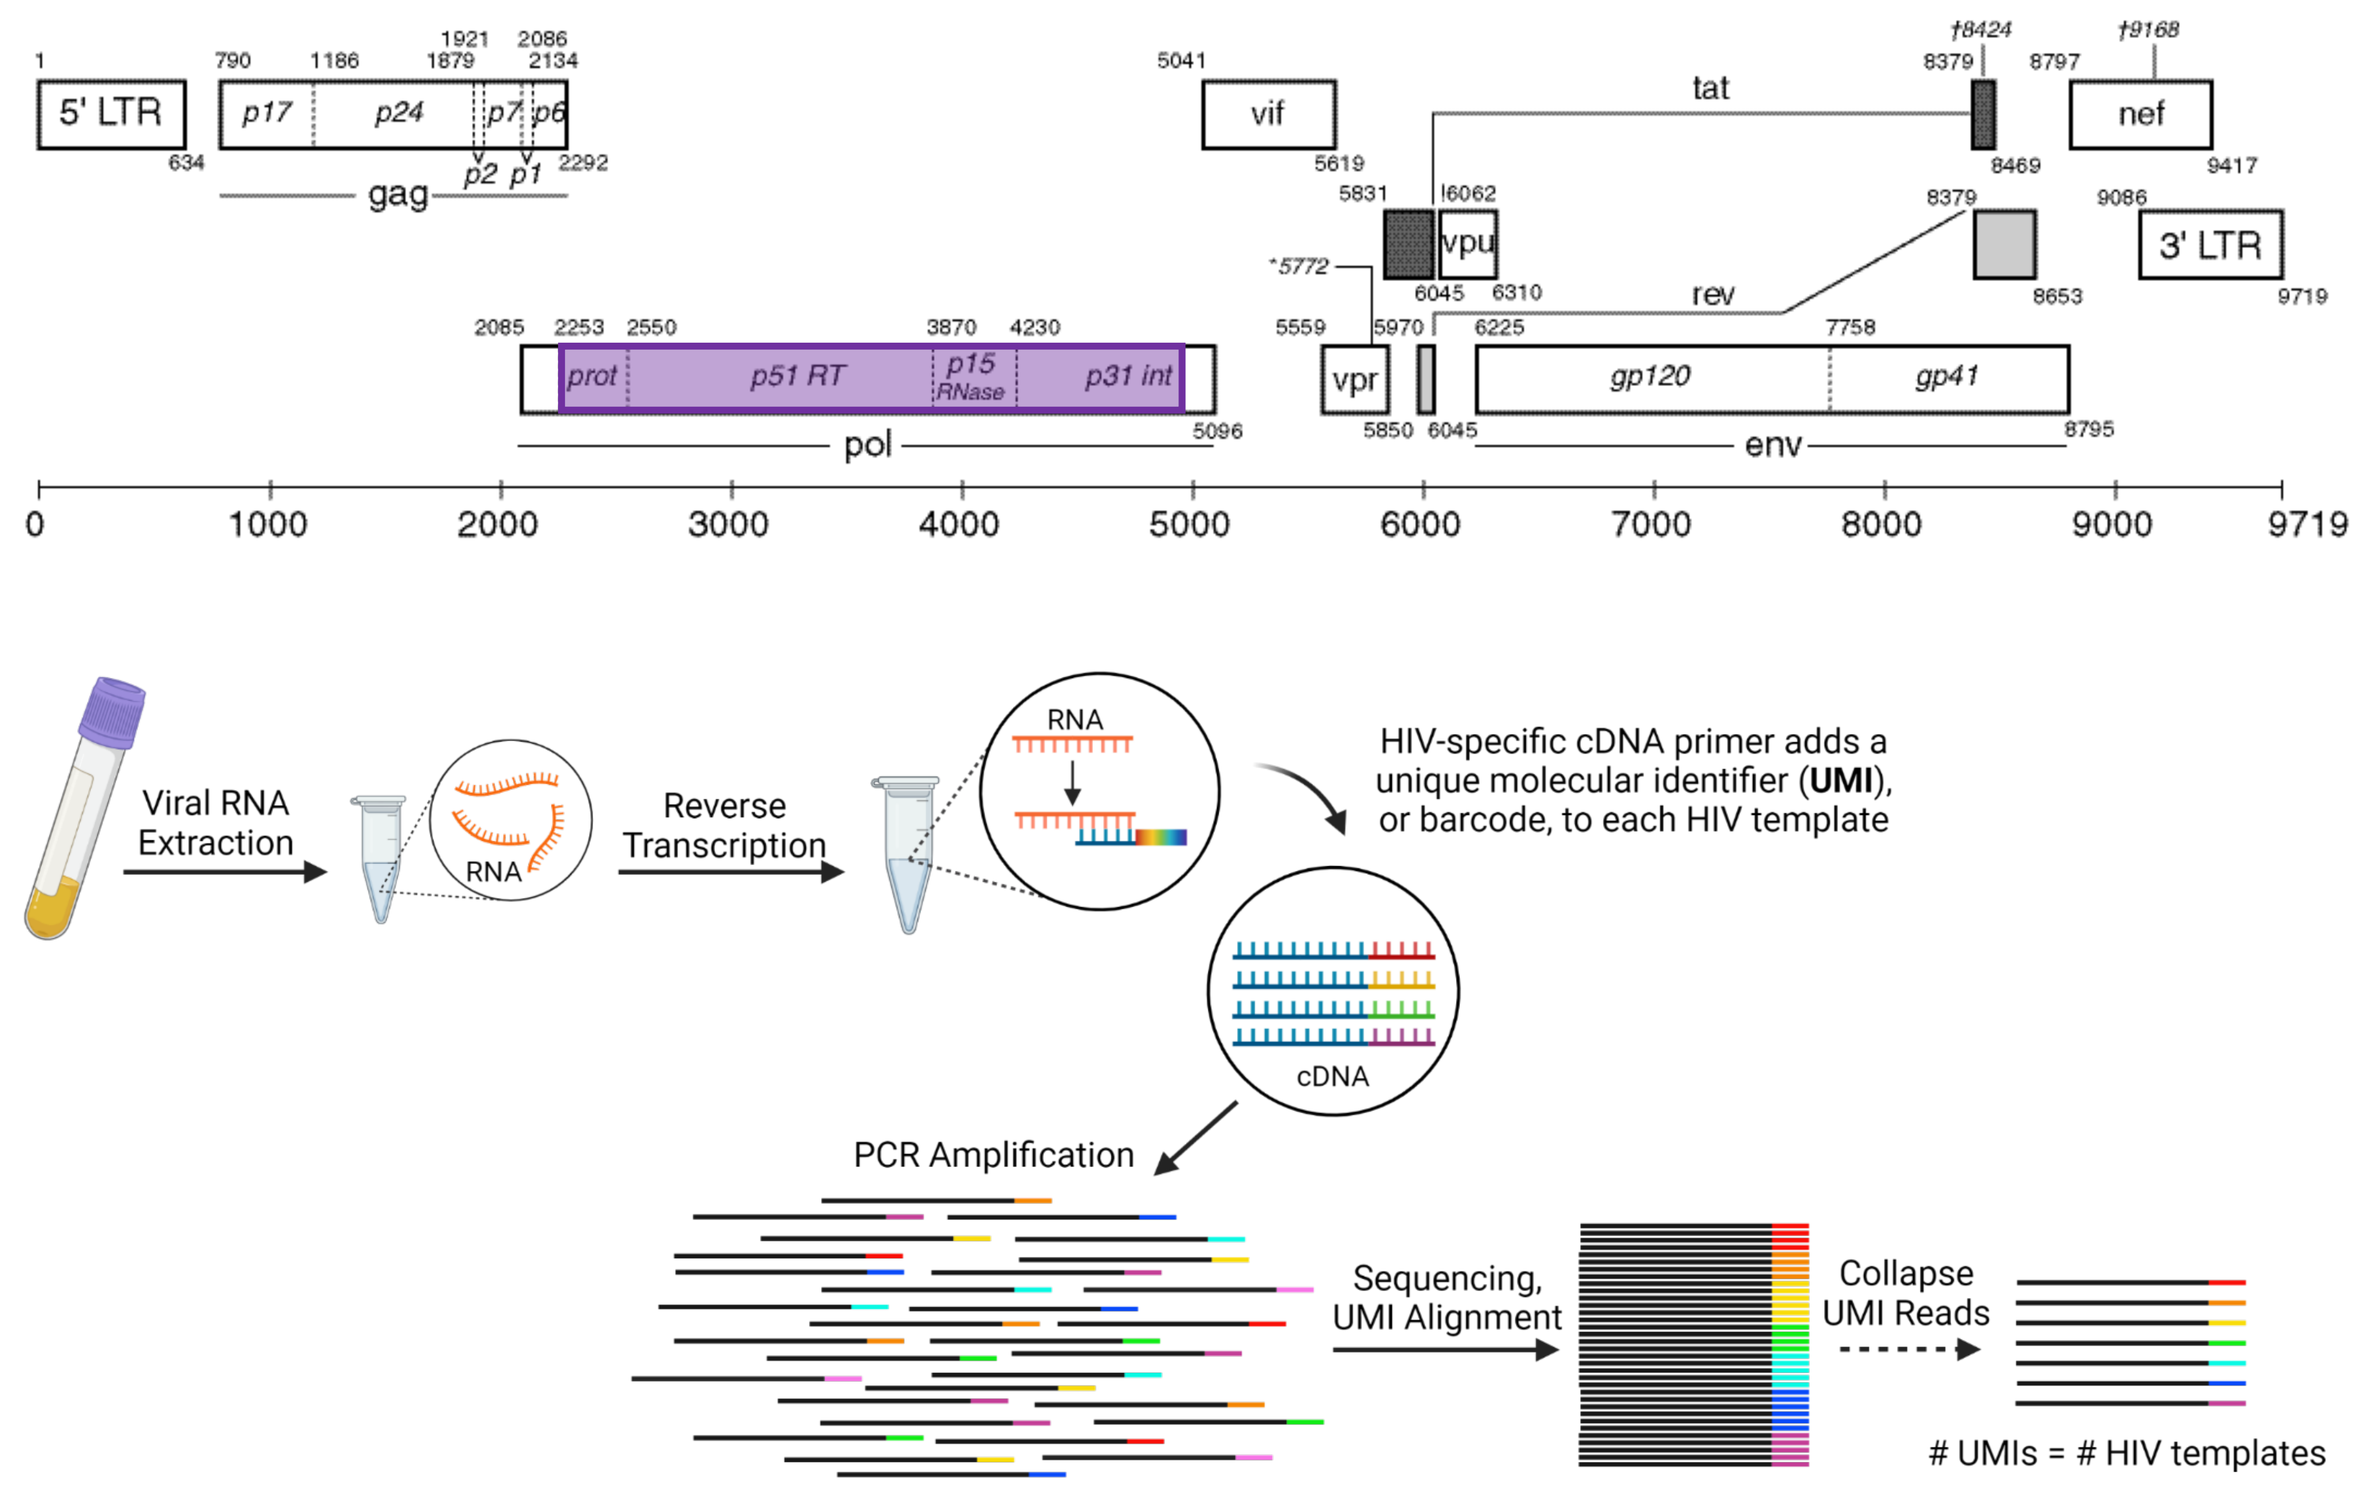
5' LTR
p17
p24
p7
p6
p2
p1
gag
vif
tat
nef
vpu
vpr
rev
3' LTR
prot
p51 RT
p15 RNase
p31 int
pol
gp120
gp41
env
Viral RNA Extraction
RNA
Reverse Transcription
HIV-specific cDNA primer adds a unique molecular identifier (UMI), or barcode, to each HIV template
cDNA
PCR Amplification
Sequencing, UMI Alignment
Collapse UMI Reads
UMIs = # HIV templates

Figure 1: PacBio HIV genotyping assay design. Assay targets most of HIV *pol* (codons PR19 to IN270) and adds barcodes (UMIs) during reverse transcription which are used to quantify the number of plasma HIV RNA templates sequenced and calculate mutational frequency. We aimed to sequence ≥100 templates per specimen; however, sequencing depth was limited for specimens with lower HIV RNA loads.

- HIV drug resistance (**DR**) was assessed using Stanford HIV Database with resistance to PIs, NRTIs, and NNRTIs defined by genotypic susceptibility scores (**GSS**) ≥30 and to DTG by GSS ≥10
- Phenotyping of participant-derived recombinant viruses was performed using a single-cycle reporter assay (Smith *et al.* 2019, PMID: 30803972) to determine the DTG 50%-effective concentration (**EC_{50}**) and EC_{50} fold-change between screen/enrollment and subsequent timepoints
- We conducted analyses in SAS using generalized estimating equations to assess the association between selection of DTG resistance and viral suppression, accounting for correlations within subjects due to repeated measures over time and assuming an autoregressive correlation structure

Results

Pre-DTG Resistance & Virologic Outcome:

- Genotyping at screen/enrollment was successful for 168/181 (92.8%) participants (12 specimens unavailable and one did not amplify)
- Resistance to PI was detected in 20/163 (12.3%); to NNRTI in 83/168 (49.4%); to NRTI in 76/168 (45.2%); and to participants' OBT (≥1 OBT drug) in 67/168 (39.9%) using ≥1% mutant threshold (**Figure 2**)
- Minority variants (<20% frequency) were uncommon at screen/enrollment
- Pre-DTG INSTI mutations were detected in 10/168 (5.9%) participants, all at <15% frequency and with GSS of 10 (H51Y, E138K, G140R, S147G)

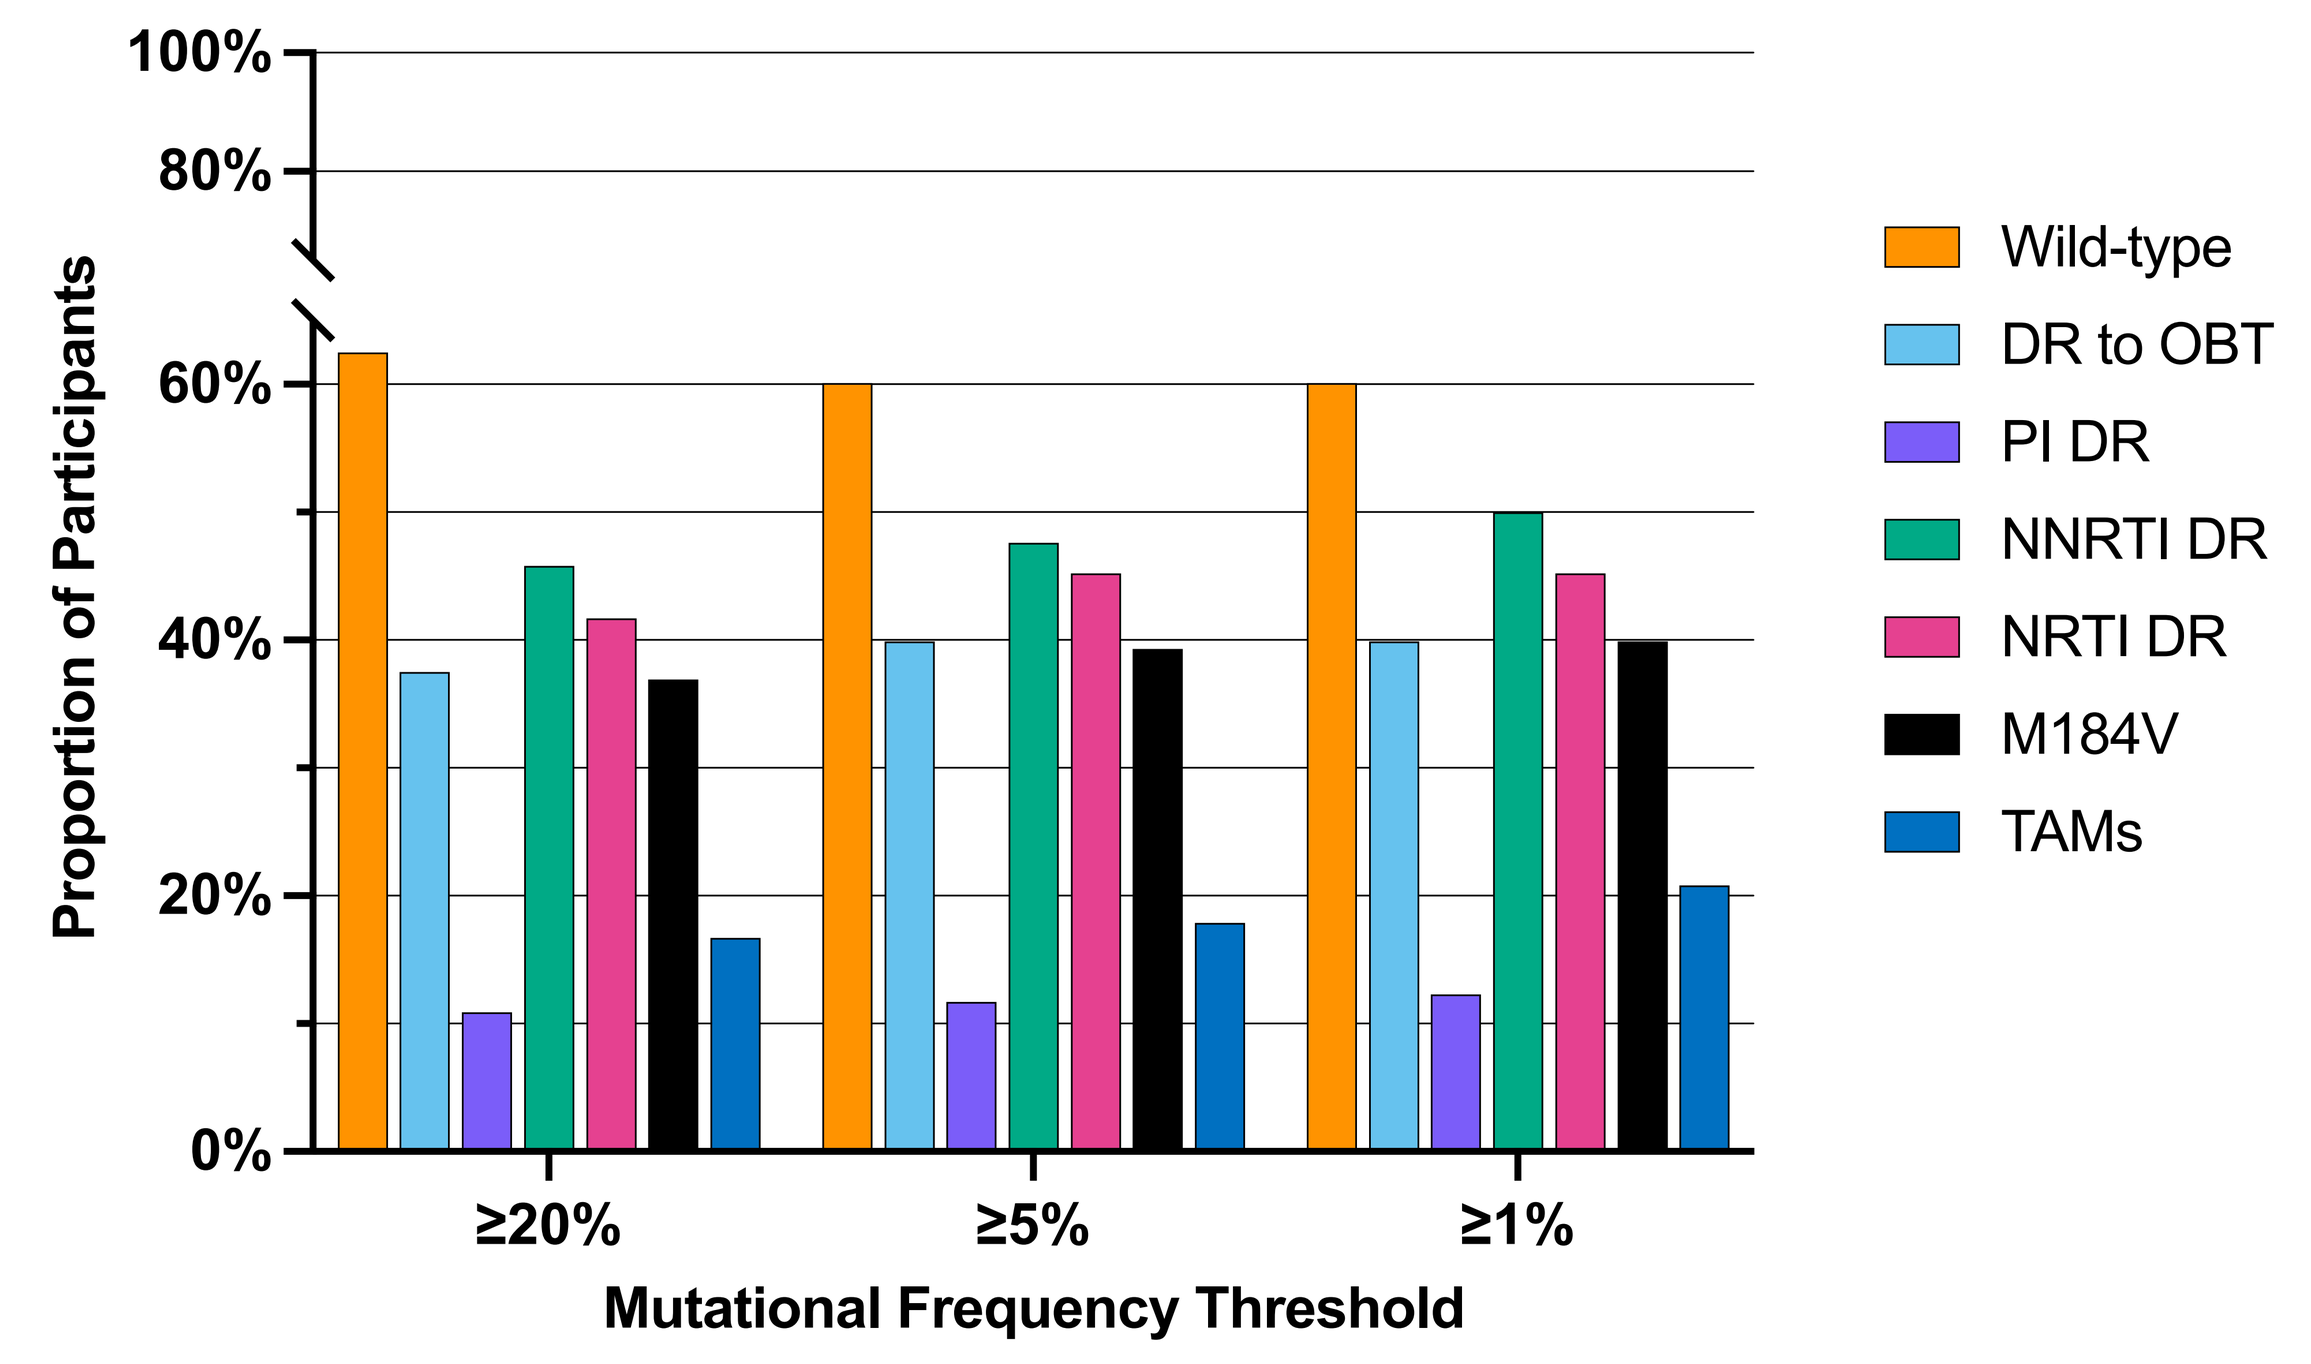
100%
80%
60%
40%
20%
0%
Proportion of Participants
≥20%
≥5%
≥1%
Mutational Frequency Threshold
Wild-type
DR to OBT
PI DR
NNRTI DR
NRTI DR
M184V
TAMs

Figure 2: Pre-DTG HIV genotypes at study screen/enrollment from 168/181 P1093 participants. Pre-DTG resistance was categorized as DR to OBT, to antiretroviral drug classes, or specific resistance pathways using mutational frequency thresholds of ≥20%, ≥5% and ≥1%. The median number of HIV templates sequenced was 162 (IQR: 92-255).

- Pre-DTG resistance was not associated with PDVF/ intermittent viremia using ≥20% (**Table 1**), ≥5%, or ≥1% mutational frequency threshold

Table 1: Pre-DTG genotypes and virologic failure/viremia

Genotype	Viremia (n=49)	Suppressed (n=120)	Total (n=169)	Risk Ratio (95% CI)
Wild-type	71	34	105	ref
DR (≥20%) to OBT	48	15	63	0.74 (0.44, 1.24)
No PI Resistance	103	44	147	ref
PI Resistance (≥20%)	14	4	18	0.74 (0.30, 1.82)
No NNRTI Resistance	63	28	91	ref
NNRTI Resistance (≥20%)	56	21	77	0.89 (0.55, 1.43)
No NRTI Resistance	68	30	98	ref
NRTI Resistance (≥20%)	51	19	70	0.89 (0.55, 1.44)
No M184V	73	33	106	ref
M184V (≥20%)	46	16	62	0.83 (0.50, 1.38)
No TAMs	97	43	140	ref
TAMs (≥20%)	22	6	28	0.70 (0.33, 1.48)

Results

Selected DTG-Associated Resistance:

- PDVF/intermittent viremia occurred in 49/181 (27.1%) participants – n=46 and n=3, respectively – with a median viral load of 5,644 c/mL (IQR: 1,469-37,891 c/mL)
- DTG-associated mutations were detected in 9/49 (18.4%) at ≥20% cut-off, 11/49 (22.5%) at ≥5% cut-off, and 12/49 (24.5%) at ≥1% cut-off (**Table 2**)
- Phenotypic analyses of 10 participants' genotypes found reduced DTG susceptibility in 7/7 participants with major INSTI mutations and similar DTG susceptibility in 3/3 participants with no or accessory only INSTI mutations compared to screen/enrollment (**Table 3**)
- Selection of DTG-associated mutations at viremia trended towards increased viral suppression <400 c/mL (OR 1.53, 95% CI 0.71-3.30, p=0.28) and was significantly associated with increased viral suppression <40 c/mL (OR=2.15, 95% CI 1.02-4.52, p=0.043) compared to no selection of DTG resistance (**Figure 3**)

Table 2: Participants with dolutegravir-associated mutations detected at PDVF/intermittent viremia

PID	Age at Entry (yrs)	Pre-DTG Genotype*	OBT	Visit Week**	Viral Load (c/mL)	Subtype	# of HIV Templates Sequenced†	Protease Genotype	Reverse Transcriptase Genotype	Integrase Genotype	Outcome after DTG Resistance Detected?	Weeks on Study
1	1.08	PR: wild-type RT: M184V	d4T+3TC	16	9,429	AE	58	none	M184V, 100%	T66I, 10.3%; G118R, 98.3%	Added LPV/r to OBT, suppressed	38
2	12.25	PR: I84V RT: wild-type	TDF+FTC	24	302	B	32	I84V, 56.2%	none	S153Y, 3.1%; R263K, 15.6%	Added RPV to OBT, did not suppress	190
3	2.64	PR: wild-type RT: M184V	ZDV+3TC	24	2,532	C	39	none	M184V, 100%; H221Y, 100%	G118R, 89.7%	Added ATV/r to OBT, suppressed	192
4	5.26	Wild-type	ZDV+3TC	29	16,627	B	140	none	M184V, 100%	E92Q, 35.0%; G118R, 27.9%; N155H, 7.1%; R263K, 3.6%	No change in ART, did not suppress	34
5	7.72	PR: wild-type RT: K103S, V106I, V108I, M184V, G190A	ZDV+3TC	48	1,408	B	8	none	K103S, 100%; M184V, 100%; G190A, 100%	H51Y, 12.5%; T97A, 62.5%; G118R, 37.5%; S147G, 62.5%; N155H, 62.5%	No change in ART, did not suppress	72
6	2.48	PR: wild-type RT: M41L, M184V	ZDV+3TC	48	5,806	B	14	none	M184V, 100%	H51Y, 28.6%; R263K‡, 21.4%	No change in ART, resuppressed but had intermittent viremia	168
7	2.44	PR: wild-type RT: M184V	ZDV+3TC	87	9,308	AE	93	none	D67N, 7.5%; M184V, 100%	E92G, 10.8%; N155H, 80.6%	No change in ART, resuppressed but then had viremia	156
8	0.19	PR: wild-type RT: Y188H	3TC+ABC	89	13,748	D	65	none	M184V, 100%	H51Y, 10.8%; G118R, 1.5%	No change in ART, suppressed	192
9	5.39	PR: wild-type RT: K103N	ABC+3TC	96	3,414	A	52	none	K103N, 100%; M184V, 53.8%	E138K, 3.9%; Q148K, 3.9%	No change in ART, resuppressed but then had viremia	180
10	0.80	Wild-type	ZDV+3TC	139	341	F	2	none	M184V, 100%	T66I, 100%; G118R, 100%§ E138A, 100%	Did not suppress, but limited additional follow-up; study team recommended participant discontinue study due to DTG resistance	144
11	4.04	PR: wild-type RT: K103N, M184V	ZDV+3TC	144	1,622	A	47	M46L, 2.1%	K103N, 100%; M184V, 100%	G140E, 2.1%; R263K, 48.9%	No change in ART, suppressed	180
12	16.95	PR: M46L RT: M41L	TDF+FTC +EFV	192	555	B	10	none	M41L, 100%	L74M, 100%; G118R, 100%	No additional follow-up, final study visit	192

*All participants were treatment-experienced
**Visit week of first plasma specimen with dolutegravir resistance detected
†Median number of HIV templates sequenced from viremic specimens was 58 (IQR: 10-140)
‡R263K was not detected in subsequent viremic specimens at weeks 84 and 132
§G118R was detected in plasma with HIV RNA=175 c/mL by Sanger, but plasma did not amplify by PacBio; note, 16/18 viremic plasma with HIV RNA <200 c/mL amplified by Sanger & 4/18 by PacBio

Table 3: Phenotypic analyses of DTG-resistant and DTG-susceptible genotypes

PID	Country	Subtype	Week[a]	INSTI resistance mutations	RC[b]	Dolutegravir EC_{50} (nM)[c]	FC[d]	Bictegravir EC_{50} (nM)[c]	FC[d]
1	Thailand	AE	0	L74I	95	1.7 ± 0.18		2.1 ± 0.49	
			20	T66I, L74I, G118R	29	**32 ± 10**	**19**	**6.8 ± 1.4**	**3.2**
2	USA	B	0	none					
			162	E138T, S147G, R263K	55	**12 ± 2.9**[f]	**5.0**[g]	**14 ± 1.5**[f]	**5.8**[g]
3	South Africa	C	0	none	80	1.7 ± 0.22		1.8 ± 0.67	
			32	L74I, G118R	35	**18 ± 6.0**	**11**	**8.2 ± 2.6**	**4.6**
			48	G118R	21	**46 ± 18**	**27**	**10 ± 2.3**	**5.6**
			48	T97A, G118R	17	**110 ± 78**	**65**	**20 ± 9.9**	**11**
5	Brazil	B	0	none					
			51	G118R, E138K, V151I	71	**19 ± 1.9**[f]	**7.9**[g]	**8.7 ± 2.9**[f]	**3.6**[g]
9	Kenya	A	0	none	25	2.4 ± 0.73		1.8 ± 0.23	
			96	E138K, Q148K	47	**58 ± 11**	**24**	**55 ± 17**	**31**
10	Brazil	F	0	none	33	3.2 ± 1.2		2.7 ± 0.88	
			139	T66I, G118R, E138A	13	**52 ± 26**	**16**	**14 ± 4.0**	**5.2**
11	Kenya	A	0	none	88	1.5 ± 0.28		2.1 ± 0.070	
			144	R263K	58	**5.5 ± 0.92**[e]	**3.7**	4.8 ± 0.94	2.3
13	Botswana	C	0	none	72	2.0 ± 0.37		2.2 ± 0.15	
			36	none	122	2.7 ± 1.1	1.4	2.3 ± 1.1	1.0
14	Thailand	AE	0	none	56	2.4 ± 0.63		2.2 ± 0.49	
			56	none	86	2.1 ± 0.80	0.88	2.5 ± 0.79	1.1
15	Kenya	AG	0	none	82	2.5 ± 0.32		2.7 ± 0.37	
			91	G140R	6.3	UTD[h]	UTD	UTD	UTD
			96	E157Q	33	4.1 ± 1.3	1.6	5.0 ± 1.0	1.9

[a] Weeks of dolutegravir treatment
[b] Replication capacity, expressed as the % of $HIV\text{-}1_{NL4\text{-}3}$ titer
[c] Mean ± standard deviation
[d] Fold change in EC_{50} value relative to the corresponding week-0 clone, unless otherwise indicated
[e] Bold type indicates a significant difference compared to the EC_{50} for the corresponding week-0 clone (P < 0.05, ANOVA with Tukey's multiple comparisons test), unless otherwise indicated
[f] Bold type indicates significant difference compared to EC_{50} for $HIV\text{-}1_{NL4\text{-}3}$ (P < 0.05, ANOVA with Tukey's multiple comparisons test)
[g] Fold change relative to $HIV\text{-}1_{NL4\text{-}3}$ (EC_{50} for dolutegravir, 2.4 ± 0.72 nM; EC_{50} for bictegravir, 2.4 ± 0.67nM)
[h] UTD, unable to determine due to insufficient replication capacity

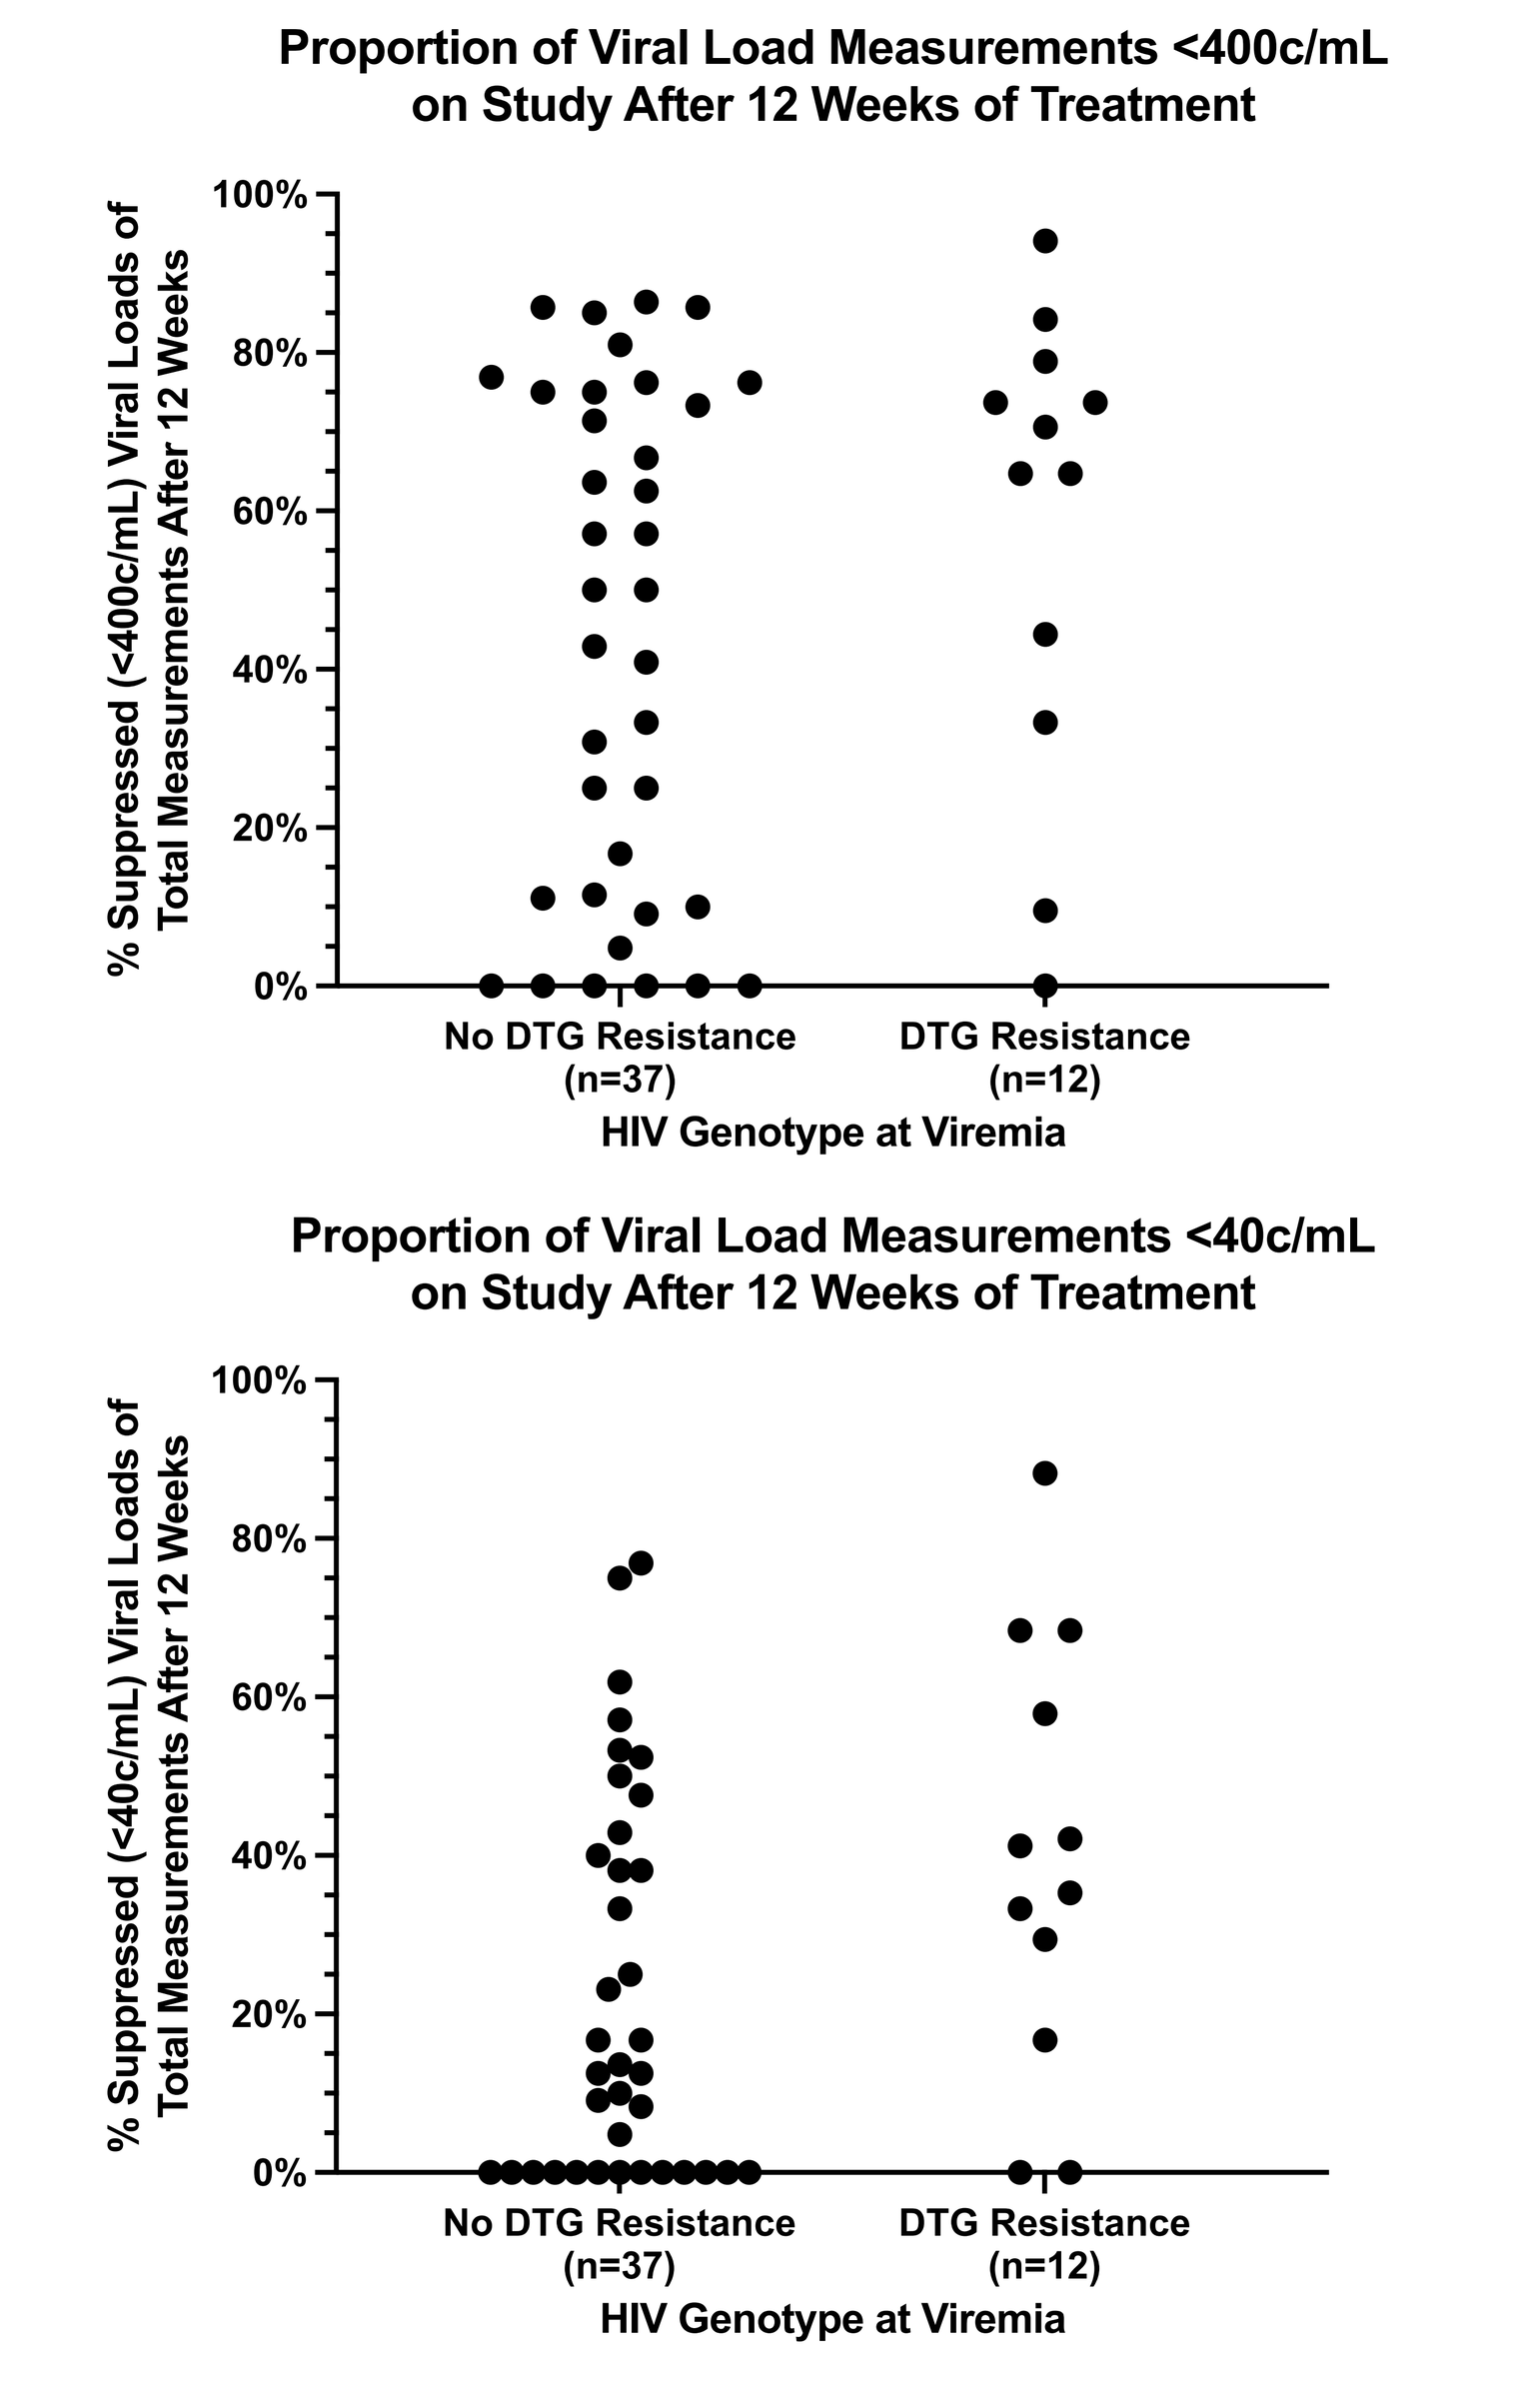
Proportion of Viral Load Measurements <400c/mL on Study After 12 Weeks of Treatment
% Suppressed (<400c/mL) Viral Loads of Total Measurements After 12 Weeks
No DTG Resistance (n=37)
DTG Resistance (n=12)
HIV Genotype at Viremia
Proportion of Viral Load Measurements <40c/mL on Study After 12 Weeks of Treatment
% Suppressed (<40c/mL) Viral Loads of Total Measurements After 12 Weeks
No DTG Resistance (n=37)
DTG Resistance (n=12)
HIV Genotype at Viremia

Figure 3: Proportion of suppressed viral loads over study period in participants by genotype at viremia. Dots reflect proportion of total viral load measurements below suppression threshold for each participant, where viral suppression was defined as <400 c/mL (top) or <40 c/mL (bottom).

Summary

- Resistance at screen/enrollment to one or more drugs included as OBT was common in this cohort and was not associated with PDVF/intermittent viremia on DTG-based ART
- Plasma from PDVF/intermittent viremia detected selected resistance to DTG in ~25%, including as minority variants either alone or in addition to majority variants
- Selection of DTG resistance was significantly associated with increased viral suppression compared to no DTG resistance at PDVF/intermittent viremia; suggesting that intermittent adherence and/or low-level viremia – as observed in PID9 (**Figure 4**) – contributes to the selection of DTG-associated mutations
- Phenotypic analyses of PDVF/intermittent viremias detected resistance at levels predicted by genotypes; interestingly, bictegravir susceptibility appeared less impacted by the same INSTI mutations compared to DTG
- Analyses assessing all potential covariates predictive of DTG-associated mutation selection (e.g., enrollment viral load, CD4%, viral load pattern, adherence, dose and formulation of DTG, etc.) are ongoing

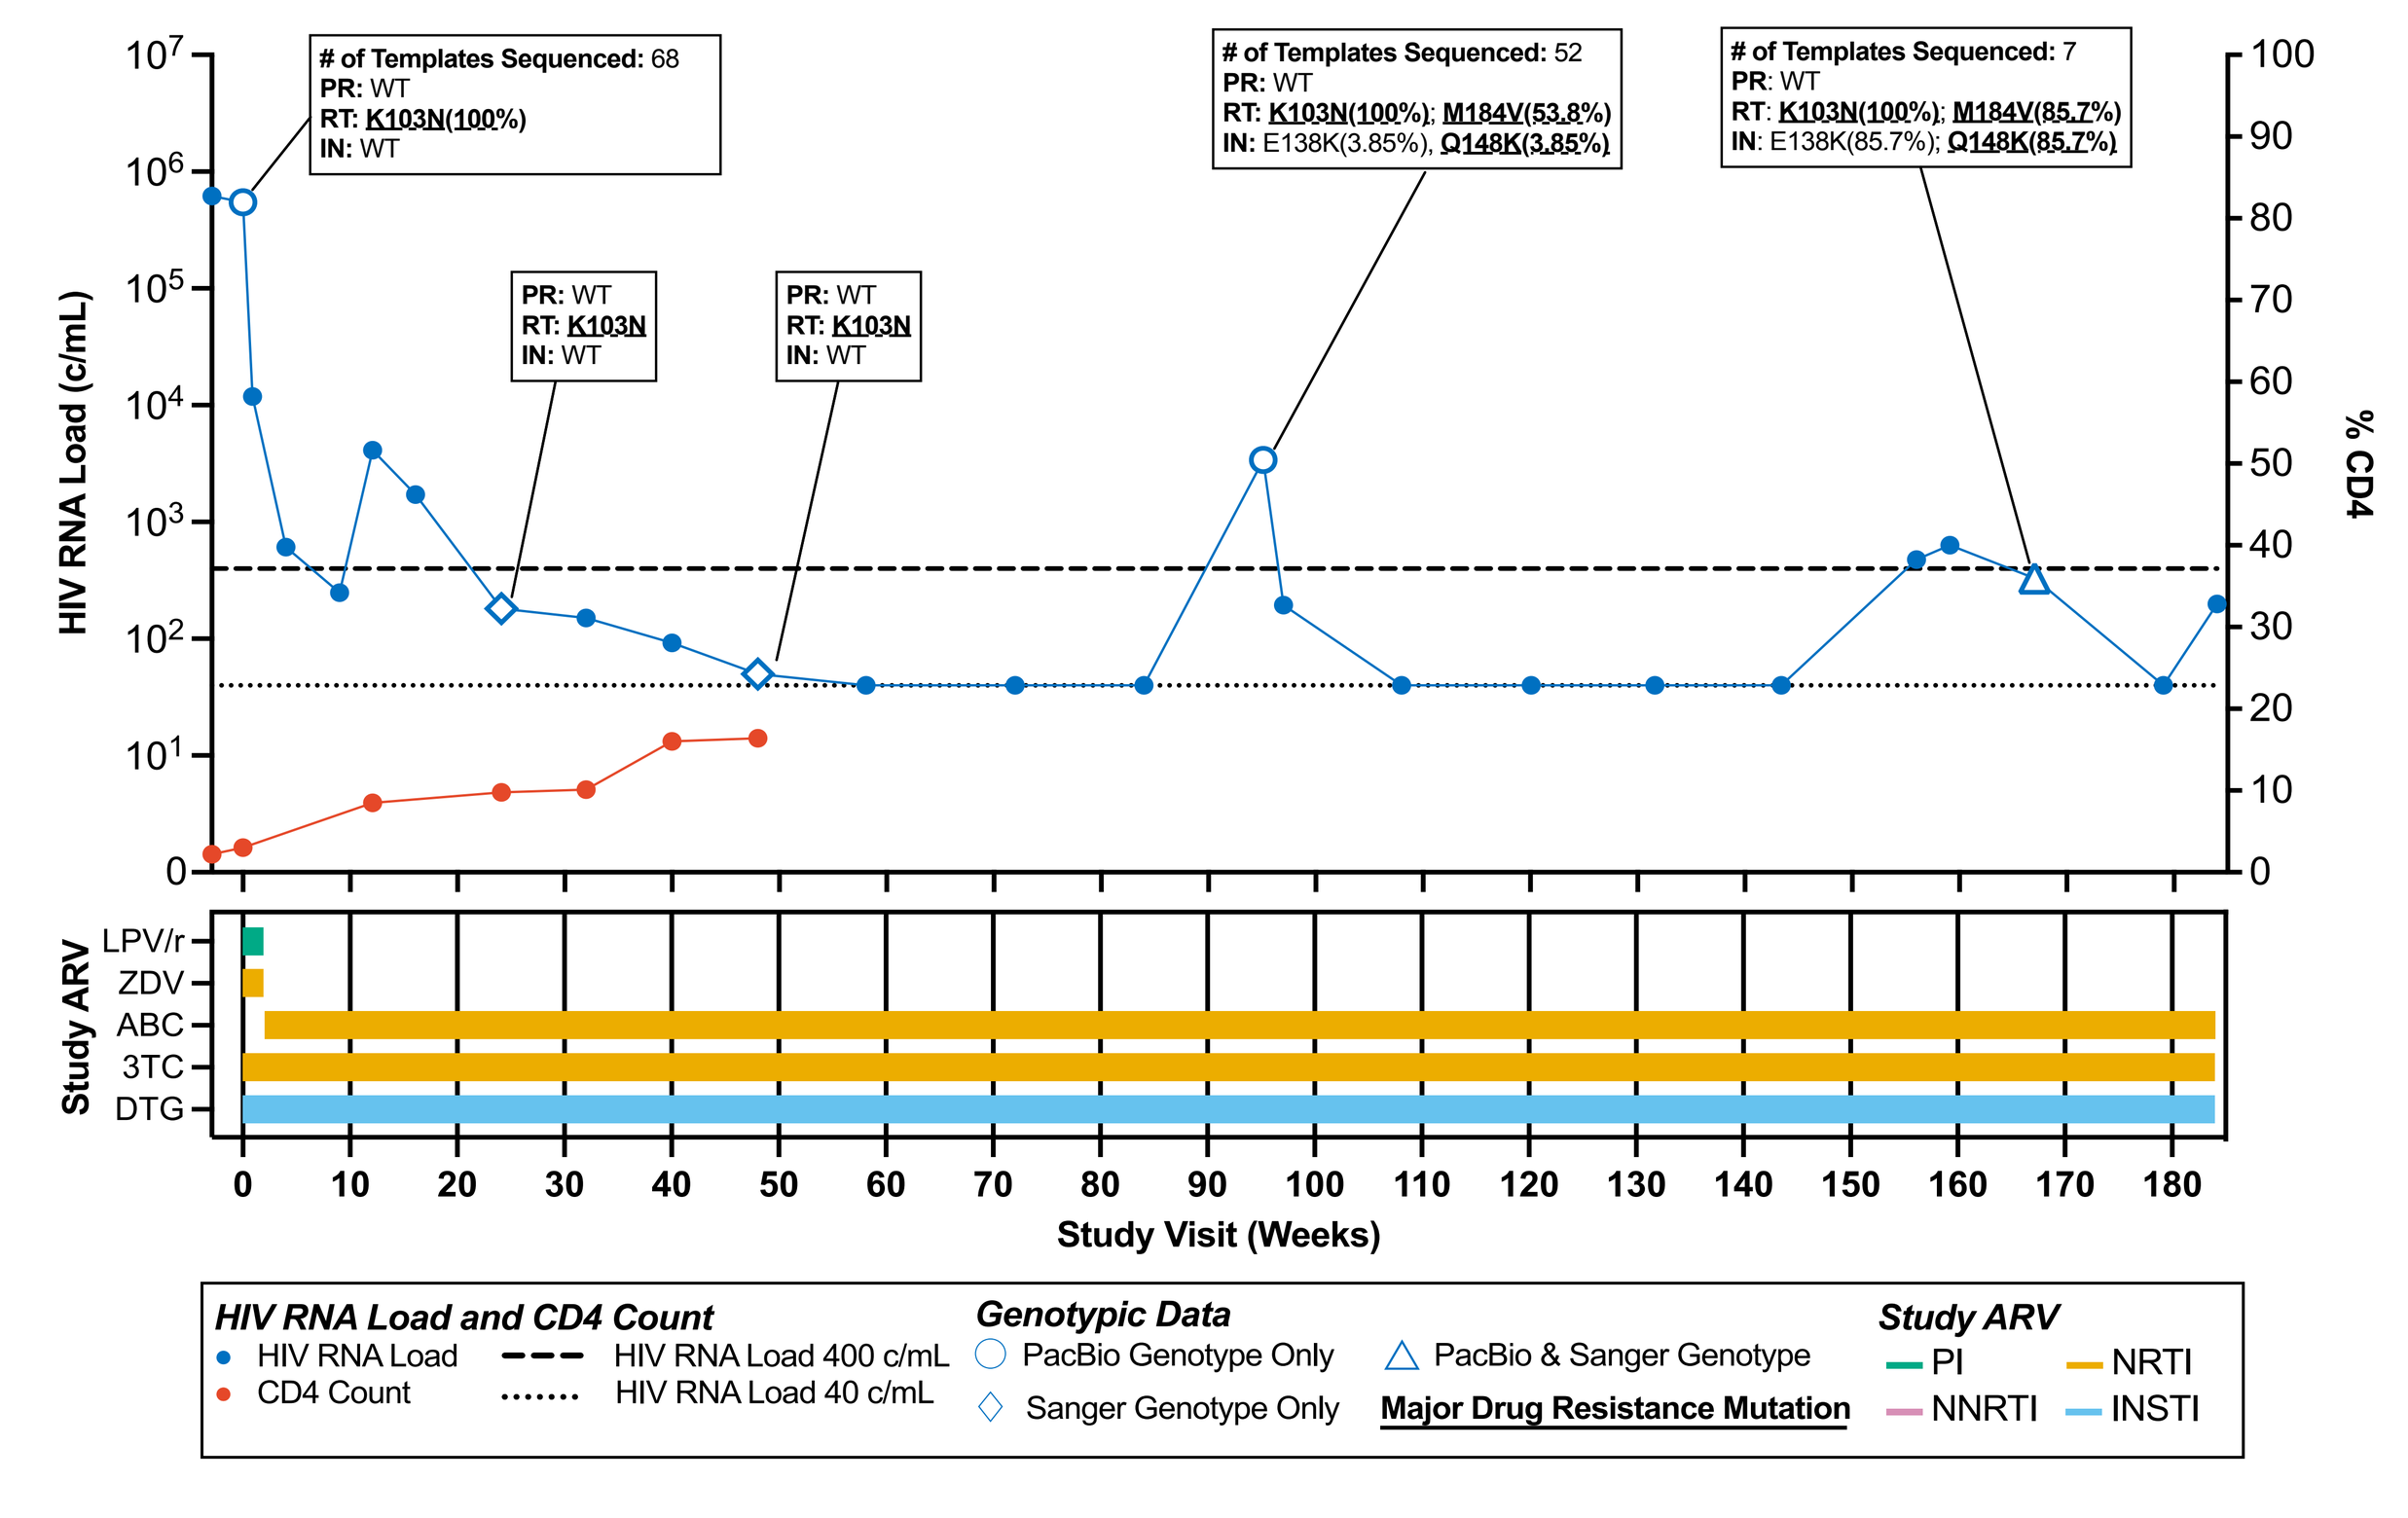
of Templates Sequenced: 68
PR: WT
RT: K103N(100%)
IN: WT
PR: WT
RT: K103N
IN: WT
PR: WT
RT: K103N
IN: WT
of Templates Sequenced: 52
PR: WT
RT: K103N(100%); M184V(53.8%)
IN: E138K(3.85%), Q148K(3.85%)
of Templates Sequenced: 7
PR: WT
RT: K103N(100%); M184V(85.7%)
IN: E138K(85.7%); Q148K(85.7%)
HIV RNA Load (c/mL)
% CD4
Study ARV
LPV/r
ZDV
ABC
3TC
DTG
Study Visit (Weeks)
HIV RNA Load and CD4 Count
HIV RNA Load
CD4 Count
HIV RNA Load 400 c/mL
HIV RNA Load 40 c/mL
Genotypic Data
PacBio Genotype Only
Sanger Genotype Only
PacBio & Sanger Genotype
Major Drug Resistance Mutation
Study ARV
PI
NNRTI
NRTI
INSTI

Figure 4: Viral load, CD4%, genotype, and treatment regimen on study for PID9

Acknowledgements

The authors gratefully acknowledge the dedication and commitment of the 181 study participants and their families without whom this study would not have been possible. The authors also wish to acknowledge the entire IMPAACT 1093 Protocol team, NIAID, NICHD, and the twenty-five IMPAACT sites and staff for their contributions to this work.

IMPAACT
International Maternal Pediatric Adolescent AIDS Clinical Trials Network

Overall support for the International Maternal Pediatric Adolescent AIDS Clinical Trials Network (IMPAACT) was provided by the National Institute of Allergy and Infectious Diseases (NIAID) with co-funding from the Eunice Kennedy Shriver National Institute of Child Health and Human Development (NICHD) and the National Institute of Mental Health (NIMH), all components of the National Institutes of Health (NIH), under Award Numbers UM1AI068632 (IMPAACT LOC), UM1AI068616 (IMPAACT SDMC) and UM1AI106716 (IMPAACT LC), and by NICHD contract number HHSN275201800001I. The content is solely the responsibility of the authors and does not necessarily represent the official views of the NIH.

IAS 2023
23–26 July

Presented at IAS 2023, the 12th IAS Conference on HIV Science